Supercritical Anti-solvent Techniques to develop Micro-particulate Protein Delivery Systems

Sara M Haider[1], Stefan Soloman[1], Erdal Cevhar[1], Rachel H Bridson[2], Keith Smith[3], Oya H Alpar[1].
[1]School of Pharmacy, University of London, 29-39 Brunswick Square, London United, Kingdom, WC1N1AX. [2] Centre for Formulation Engineering, Department of Chemical Engineering, University of Birmingham, United Kingdom, B15 2TT.
[3]GlaxoSmithKline, New Frontiers Business Park, Harlow, United Kingdom, CM19 5AW.
Contact details: E-mail: sara.haider@pharmacy.ac.uk Tel: 00442077534888.

Abstract summary:

Protein and polymer particles were prepared using supercritical fluid (SCF) anti-solvent techniques. The particle size, morphology and residual solvent content of the powders were studied. Micron sized particles were generated using the SCF anti-solvent systems. Protein was also precipitated from the systems and production of composite particles is now underway.

Introduction:

Supercritical Fluid (SCF) technology has been shown to be a viable option in the formulation of particulate drug delivery systems such as micro- / nano-particles which control drug delivery and enhance drug stability[1].

Therapeutic proteins are increasingly being introduced to the pharmaceutical market and thus particle-engineering techniques that ensure and preserve their stability are becoming increasingly vital. Traditional drying processes such as freeze or spray drying apply potentially harmful stresses on them[2]. Compared to conventional particle formation techniques, SCF technology allows the use of mild processing conditions, environmentally benign non-toxic solvents, minimal organic solvent use, and production of particles with controllable size distributions and morphology[3,4].

Two major supercritical techniques have been developed for the production of solid particles. The first is known as Rapid Expansion of Supercritical Solutions (RESS) and involves preparing a solution of the material in the SCF and expanding it through a nozzle[5]. Due to the limited solubility of many substances in supercritical CO_2 (SC CO_2 - the most widely used SCF), anti-solvent techniques have gained favour. These involve contacting an organic solution with SC CO_2, resulting in expansion of the solution and supersaturation. This is turn leads to precipitation of the previously dissolved substance, often in the form of micro or nano particles[6]. Various anti-solvent techniques exist, differing in the mechanism by which the SCF and solution are contacted.

Using SCF techniques the aim of the current project is to form polymeric micro- and nano-particles containing vaccine antigens and to develop a single-step process to produce such particles for controlled release applications.

Method:

Solubility studies were conducted to investigate the solubility of the polymers poly(methyl methacrylate), polycaprolactone, poly(lactic acid) and poly(lactic-co-glycolic acid) in SC CO_2. The studies showed very limited solubility of the polymers in SC CO_2, leading to the decision to proceed with particle preparation via supercritical anti-solvent techniques.

Two different anti-solvent techniques have been investigated, that can be distinguished by the nozzle configurations used in each. The first (Process 1) consists of a co-axial nozzle in which the fluids are simultaneously introduced into the precipitation chamber. The second (Process 2) involves spraying a solution into a T-piece through which SC CO_2 flows.

To aid process development, the nozzle systems have first been used to produce single component particles of L-PLA and bovine serum albumin (BSA) alone. Both techniques involve using high pressure carbon dioxide to generate particles from solutions. The polymer particles are generated from organic solution, and the protein particles from aqueous solution. The latter requires modification of the SC CO_2 with ethanol.

Results:

Particle size distributions were determined using laser diffraction (Mastersizer, Malvern). It was found that both systems produced microparticles of L-PLA with mean volume diameters ranging between 5-7 micrometers (µm). However, scanning electron micrographs (SEMs) showed that the particles produced using Process 1 were not only smaller, but also more spherical and consistent in shape.

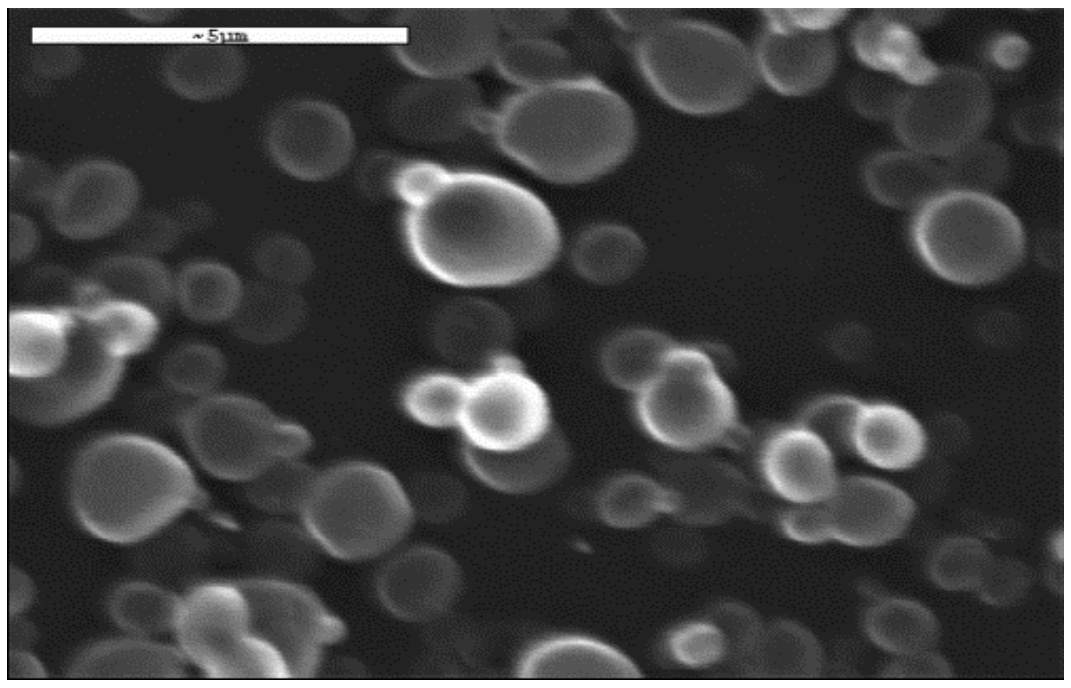
~5µm

Figure 1. SEM of PLLA microparticles produced at 200 bar and 45ºC from a 50% w/v polymer solution using Process 1.

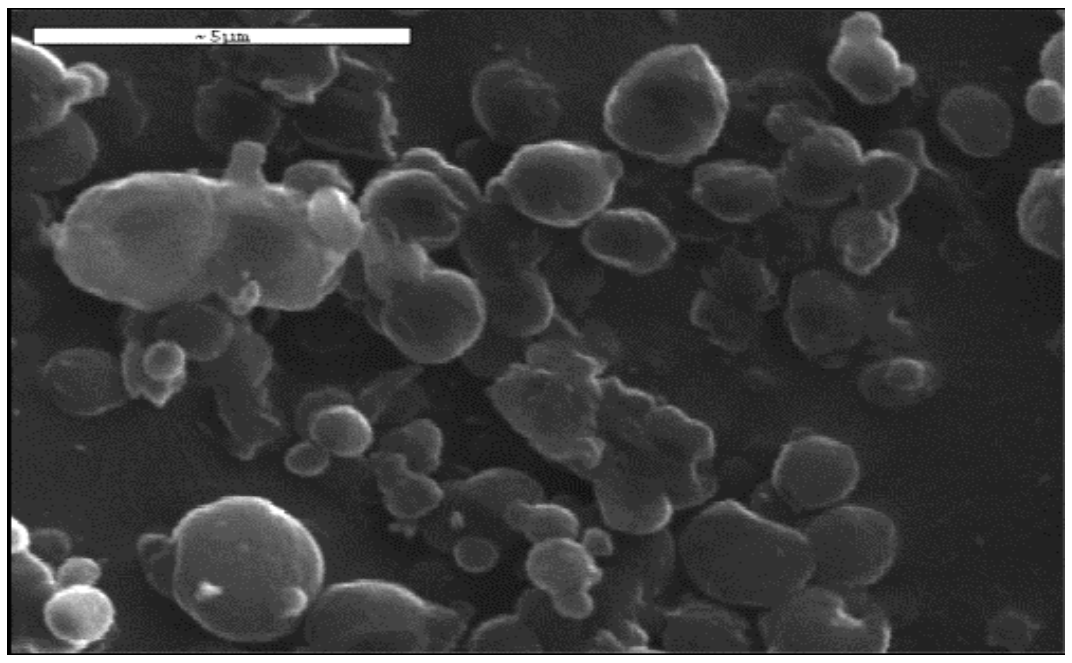
~ 5μm

Figure 2. SEM of PLLA microparticles produced at 200 bar and 45ºC from a 50% w/v polymer solution using Process 2.

The SEMs show that most particles are less than 3µm in diameter. It is suggested that the laser diffraction results were affected by the presence of aggregated particles, whereas scanning electron microscopy allowed identification of primary particles.

It was seen that size of the polymer particles could be reduced through alteration of pressure, temperature and concentration of organic polymer solution. It was seen that smaller particles were generated at higher pressures, higher temperatures and by precipitation from solutions of higher concentrations.

BSA could also be precipitated using the anti-solvent processes, but some residual ethanol was evident. Thermogravimetric analysis (TGA) was therefore used to identify the amount of residual solvent in the protein particles. Table 1 shows the mass loss which took place between 40ºC and 160ºC and which was assumed to be ethanol. It was seen that the protein particles produced through Process 2 contained less residual solvent, which is known to affect the protein structure causing denaturation.

Table 1. Summary of mass loss of formulations made from processing a 5% m/v aqueous solution using the two nozzle systems at 200 bar, 45ºC with co-solvent molar ratios of 0.2 and 0.3.

Processing system	Co-solvent molar ratio	Mean mass loss from samples of protein particles (% m/m) ±s.d.
Process 1	0.2	6.09±0.18
Process 2	0.2	5.53±0.13
Process 1	0.3	7.52±0.11
Process 2	0.3	4.91±0.41

The results point to the need for further experimental work to identify the parameters by which protein particles with minimal quantities of residual solvent can be produced.

The next step is to produce BSA loaded within microparticles of L-PLA, with controllable and well-defined mean sizes and size distributions, and to evaluate the effect of process variables on the particles.

Conclusion:

Microparticles of L-PLA and BSA have been prepared. It was seen that mean particle size was controllable by varying the solution concentration of the organic solution and the pressure and temperature of the system. Further work is to be conducted to produce loaded microparticles with the desired release profiles.

References:

1) Bustami R, Chan H, Dehghani F, Foster N. Generation of Micro-Particles of Proteins for Aerosol Delivery Using High Pressure Modified Carbon Dioxide. Pharmaceutical Research: 17: 2000; 1360-1366.
2) Chen A, Pu X, Kang Y, Liao L, Yao Y, Yin G. Study of poly(L-lactide) microparticles based on supercritical CO_2. J Mater Sci: Mater Med: 18: 2000; 2339-2345.
3) Song K, Lee C, Lim J, Lee Y. Preparation of L-PLA Submicron Particles by a Continuous Supercritical Antisolvent Precipitation Process. Korean J. Chem. Eng: 19: 2002; 139-145.
4) York P. Strategies for particle design using supercritical fluid technologies. Pharm. Sci. Tech. Today 2: 1999; 430–440.
5) Vasukumar K, Bansal A. Supercritical fluid technology in pharmaceutical research. Crips: 4: 2003; 8-12.
6) Subramaniam B, Rajewski R, Snavely K. Pharmaceutical processing with supercritical carbon dioxide. Journal of Pharmaceutical Science: 86: 1997; 885-890.

Acknowledgements:

The authors thank Dave McCarthy for his technical expertise with the SEM.

Financial support from GlaxoSmithKline and the BBSRC is gratefully acknowledged.